HORIZON (OP-106): An Exploratory Analysis of Time to Next Treatment in Patients With Relapsed/Refractory Multiple Myeloma Who Received Melflufen Plus Dexamethasone

COMy
The 6th World Congress on
CONTROVERSIES IN MULTIPLE MYELOMA (COMy)

Maria-Victoria Mateos[1]; Albert Oriol[2]; Alessandra Larocca[3]; Joan Bladé[4]; Michele Cavo[5]; Paula Rodriguez-Otero[6]; Xavier Leleu[7]; John W. Hiemenz[8]; Hani Hassoun[9]; Cyrille Touzeau[10-12]; Adrián Alegre[13]; Agne Paner[14]; Christopher Maisel[15]; Amitabha Mazumder[16]; Anastasios Raptis[17]; Johan Harmenberg[18]; Stojan Zavisic[18]; Sara Thuresson[18]; Oskar Öhman[18]; Paul G. Richardson[19]

[1]Hospital Clínico Universitario de Salamanca/IBSAL/CIC, Salamanca, Spain; [2]Institut Català d'Oncologia and Institut Josep Carreras, Hospital Germans Trias i Pujol, Badalona, Spain; [3]Myeloma Unit, Division of Hematology, University of Torino, Azienda Ospedaliero – Universitaria Città della Salute e della Scienza, Torino, Italy; [4]Hematology Department, IDIBAPS, Hospital Clinic, Barcelona, Spain; [5]Seràgnoli Institute of Hematology, Bologna University School of Medicine, Bologna, Italy; [6]Clínica Universidad de Navarra, Pamplona, Spain; [7]CHU de Poitiers, Poitiers, France; [8]Division of Hematology-Oncology, Department of Medicine, University of Florida, Gainesville, FL, USA; [9]Myeloma Service, Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [10]Centre de Recherche en Cancérologie et Immunologie Nantes-Angers (CRCINA), Institut National de la Santé et de la Recherche Médicale (INSERM), Centre National de la Recherche Scientifique (CNRS), Université d'Angers, Université de Nantes, Nantes, France; [11]Site de Recherche Intégrée sur le Cancer (SIRIC), Imaging and Longitudinal Investigations to Ameliorate Decision-making (ILIAD), Nantes, France; [12]Service d'hématologie Clinique, Centre Hospitalier Universitaire, Place Alexis Ricordeau, Nantes, France; [13]Hospital Universitario La Princesa and Hospital Universitario Quironsalud, Madrid, Spain; [14]Rush University Medical Center, Chicago, IL, USA; [15]Baylor Scott & White Charles A. Sammons Cancer Center, Dallas, TX, USA; [16]The Oncology Institute of Hope and Innovation, Glendale, CA, USA; [17]Division of Hematology-Oncology, Department of Medicine, University of Pittsburgh School of Medicine, Pittsburgh, PA, USA; [18]Oncopeptides AB, Stockholm, Sweden; [19]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA

BACKGROUND

- Outcomes remain poor for patients with relapsed/refractory multiple myeloma (RRMM), despite the availability of new therapies[1,2]
- With each subsequent relapse, patients with RRMM typically have worse outcomes, including lower response rates, shorter progression-free survival (PFS) and overall survival, and decreased treatment duration.[2-4] This decline is especially true earlier in the course of the disease[4]
- Longer time to next treatment (TTNT) is indicative of disease stabilization and clinical benefit[4-6]
 - Longer TTNT has also been associated with lower costs in patients with RRMM
- Melphalan flufenamide (melflufen) is a first-in-class peptide-drug conjugate (PDC) that targets aminopeptidases and rapidly releases alkylating agents into tumor cells (**Figure 1**)[7-11]
- In the phase 2 HORIZON study, melflufen plus dexamethasone (dex) showed clinically meaningful efficacy and a manageable safety profile in patients with poor-risk, heavily pretreated RRMM[12]
 - In the intention-to-treat population (ITT; N=157), the overall response rate (ORR) was 29%, with a median duration of response of 5.5 months
 - ORR was 26% in the triple-class–refractory population (n=119) and 24% among patients with extramedullary disease (EMD; n=55)
 - The safety profile of melflufen plus dex was consistent with previous data[13] and consisted primarily of hematologic events
 - The most common grade 3/4 adverse events (AEs) were neutropenia (79%), thrombocytopenia (76%), and anemia (43%)
 - The most common grade 3/4 nonhematologic AEs included pneumonia (10%) and hypophosphatemia (5%)
- The TTNT of melflufen plus dex in patients with RRMM was first evaluated in the phase 1/2 O-12-M1 study[6]
 - In 45 patients with RRMM who had received a median of 4 prior lines of therapy, median TTNT was 7.9 months, and median PFS was 5.7 months
- Here we present the TTNT of melflufen plus dex from HORIZON, which was a more heavily pretreated population of patients with RRMM. To our knowledge, this is the first report of TTNT in a population with a median of 5 prior lines of therapy evaluated in a clinical trial

Figure 1. Melflufen Mechanism of Action

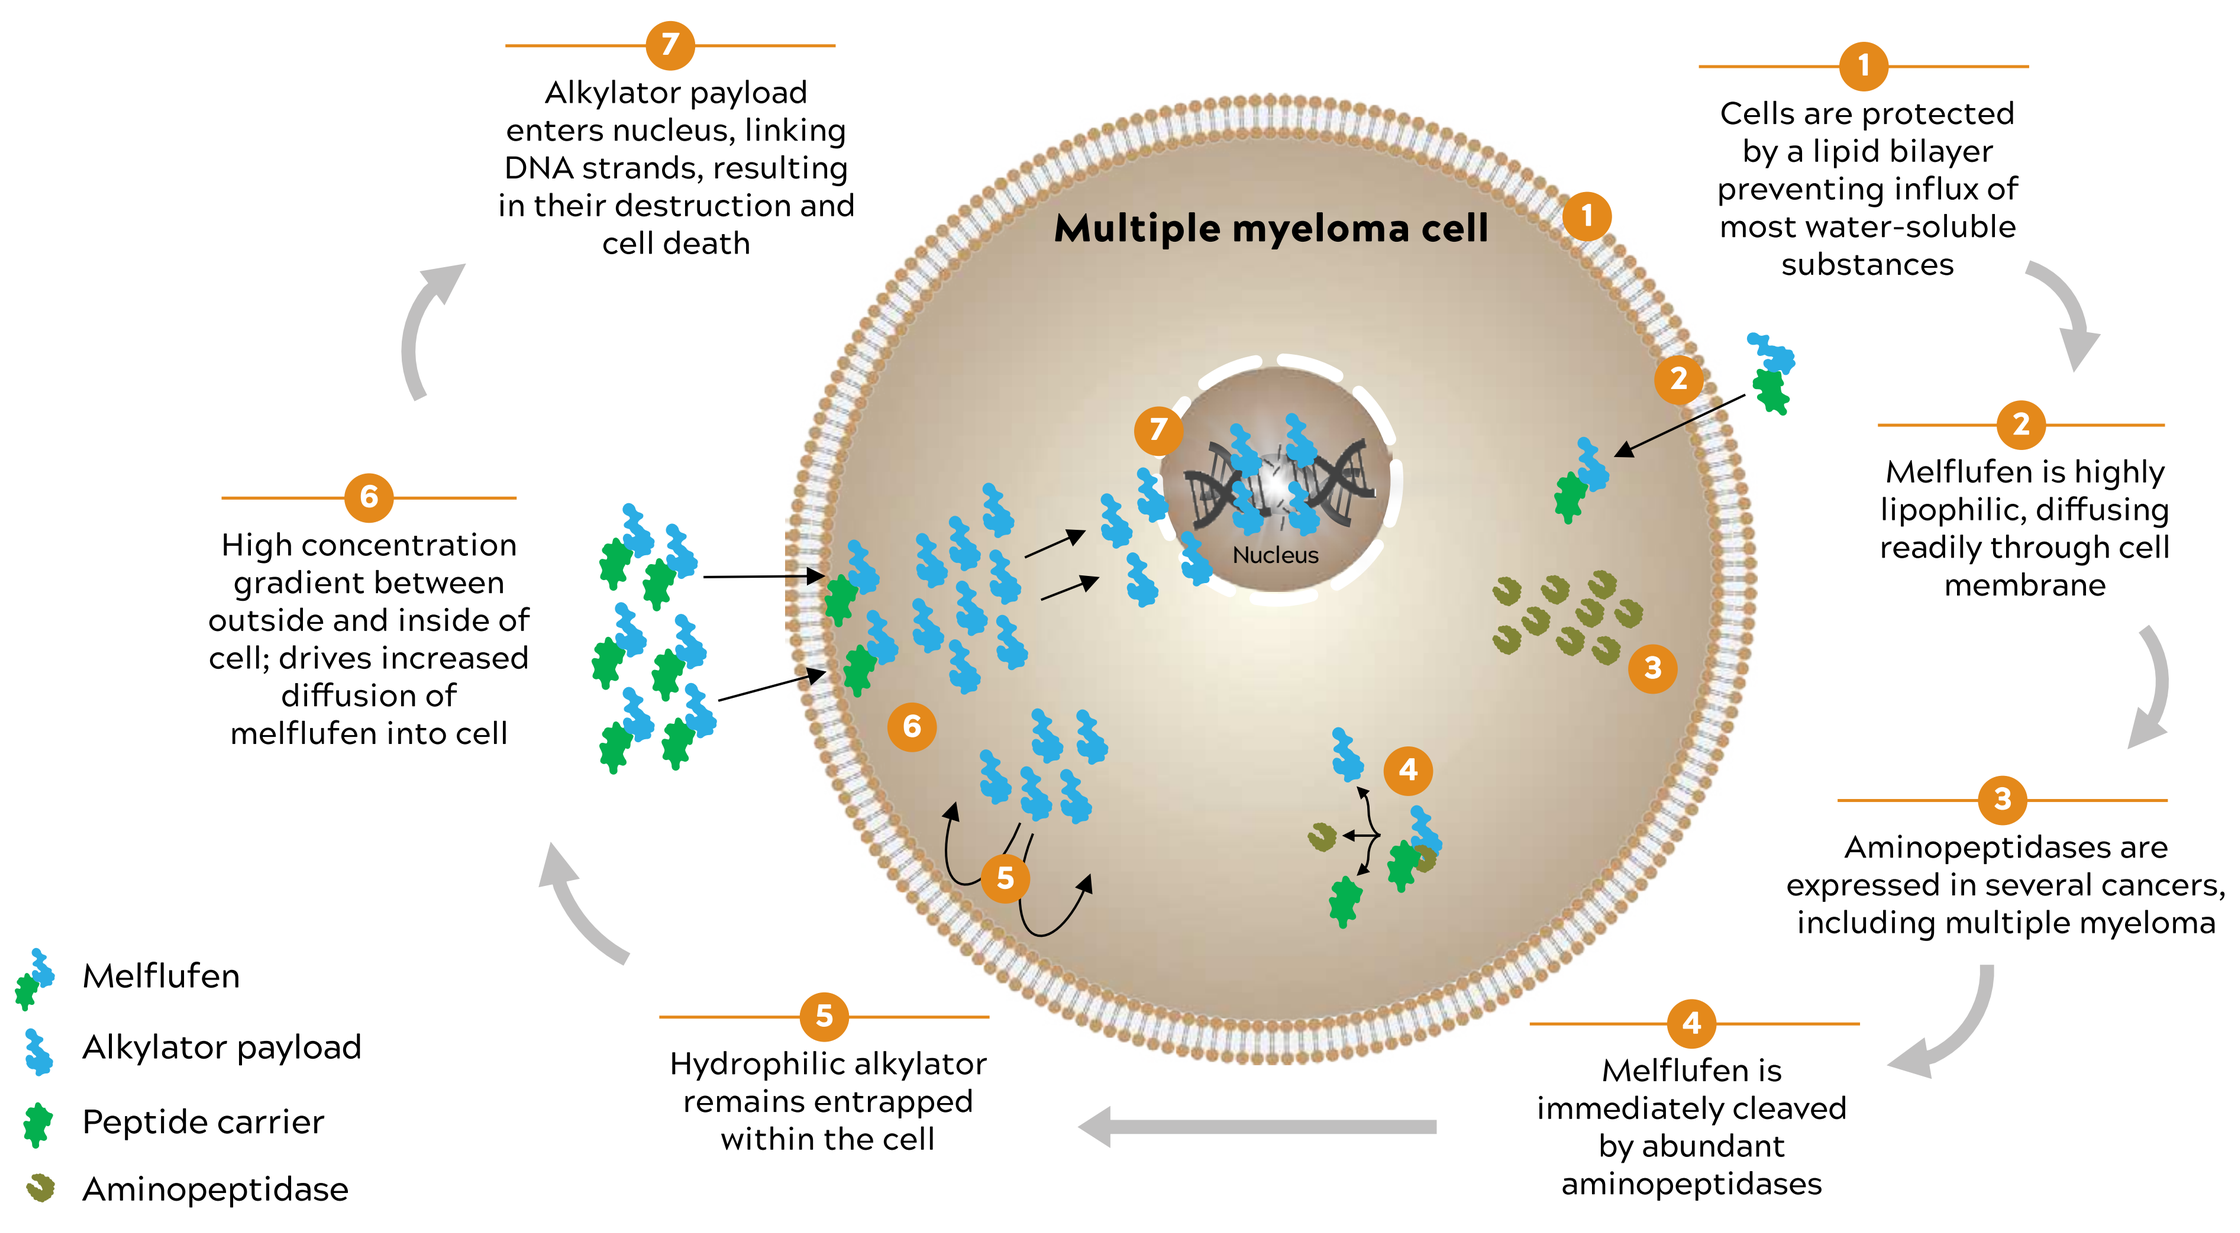
Multiple myeloma cell
Nucleus
1 Cells are protected by a lipid bilayer preventing influx of most water-soluble substances
2 Melflufen is highly lipophilic, diffusing readily through cell membrane
3 Aminopeptidases are expressed in several cancers, including multiple myeloma
4 Melflufen is immediately cleaved by abundant aminopeptidases
5 Hydrophilic alkylator remains entrapped within the cell
6 High concentration gradient between outside and inside of cell; drives increased diffusion of melflufen into cell
7 Alkylator payload enters nucleus, linking DNA strands, resulting in their destruction and cell death
Melflufen
Alkylator payload
Peptide carrier
Aminopeptidase

OBJECTIVES

- To evaluate the TTNT after melflufen plus dex in patients with RRMM in the phase 2 HORIZON study
- To describe subsequent treatments received following melflufen and treatment received prior to initiating melflufen in this heavily pretreated, poor-risk RRMM population

METHODS

- HORIZON is an ongoing pivotal, multicenter, single-arm, phase 2 study of melflufen plus dex in patients with heavily pretreated RRMM, refractory to pomalidomide (pom) and/or an anti-CD38 monoclonal antibody (mAb; **Figure 2**)
 - The primary results for the fully enrolled study have been presented[12]
- Patients received melflufen 40 mg (intravenously on day 1 of each 28-day cycle) plus dex 40 mg weekly until disease progression or unacceptable toxicity
- The primary endpoint was ORR
 - Response was assessed by the investigator per the International Myeloma Working Group uniform response criteria[14]
- TTNT was defined as the time from start of melflufen to first subsequent therapy (deaths were censored)
 - TTNT or death was defined as the time from start of melflufen to first subsequent therapy or death (whichever occurred first)
 - Patients were followed for ≤2 years after disease progression, and TTNT was retrospectively reviewed
- Subgroups of special interest included patients with triple-class–refractory disease and patients with EMD

Figure 2. Phase 2 HORIZON Study Design (NCT02963493)

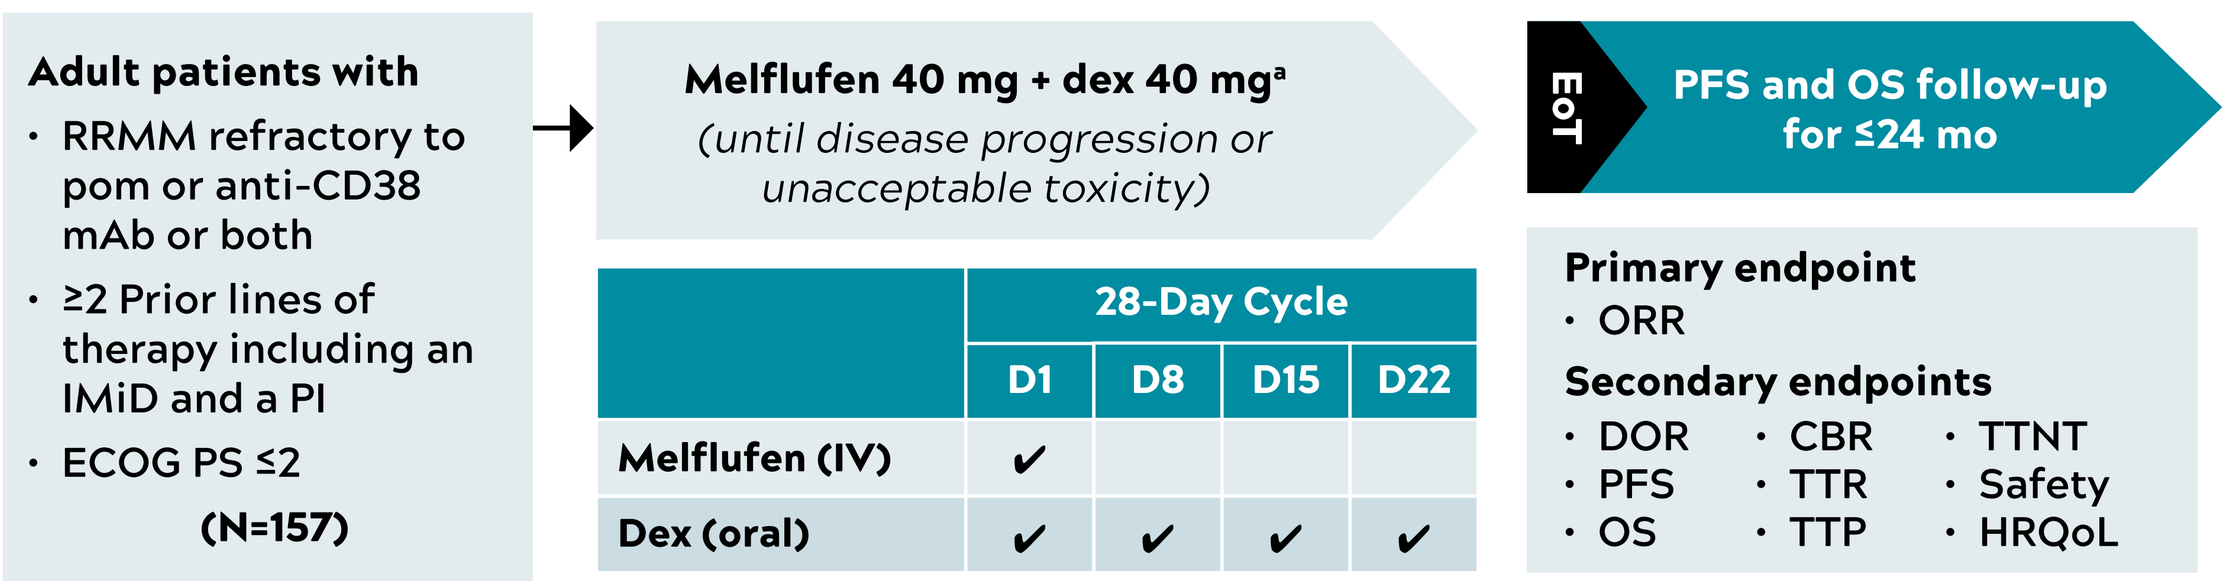
Adult patients with
• RRMM refractory to pom or anti-CD38 mAb or both
• ≥2 Prior lines of therapy including an IMiD and a PI
• ECOG PS ≤2
(N=157)
Melflufen 40 mg + dex 40 mg[a]
(until disease progression or unacceptable toxicity)
EoT
PFS and OS follow-up for ≤24 mo
28-Day Cycle
D1 D8 D15 D22
Melflufen (IV) ✔
Dex (oral) ✔ ✔ ✔ ✔
Primary endpoint
• ORR
Secondary endpoints
• DOR • CBR • TTNT
• PFS • TTR • Safety
• OS • TTP • HRQoL

[a]Patients aged >75 years received dex 20 mg.
CBR, clinical benefit rate; dex, dexamethasone; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EoT, end of treatment; HRQoL, health-related quality of life; IV, intravenous; mAb, monoclonal antibody; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PI, proteasome inhibitor; RRMM, relapsed/refractory multiple myeloma; TTNT, time to next treatment; TTP, time to progression; TTR, time to response.

RESULTS

- As of January 14, 2020 the study was fully enrolled with 157 patients, all of whom received ≥1 dose of study medication; 131 patients (83%) discontinued therapy and 26 patients (17%) remained on treatment (**Table 1**)
 - The most common primary reasons for treatment discontinuation were disease progression (56%) and AEs (17%)
- Baseline patient characteristics are shown in **Table 2**

Table 1. Patient Disposition

	ITT (N=157)	Triple-Class–Refractory (n=119)	EMD (n=55)
Discontinued therapy, n (%)	131 (83)	102 (86)	50 (91)
Primary reason for discontinuation, n (%)			
Disease progression	88 (56)	71 (60)	35 (64)
AEs	26 (17)	16 (13)	9 (16)
Patient withdrawal	7 (4)	6 (5)	0
Lack of efficacy	5 (3)	5 (4)	3 (5)
Physician choice	5 (3)	4 (3)	3 (5)
Ongoing, n (%)[a]	26 (17)	17 (14)	5 (9)

[a]Data cutoff date, January 14, 2020.
AE, adverse event; EMD, extramedullary disease; ITT, intention-to-treat.

RESULTS (CONTINUED)

Table 2. Baseline Patient Characteristics

Characteristic	ITT (N=157)	Triple-Class–Refractory (n=119)	EMD (n=55)
Median age (range), years	65 (35-86)	65 (35-86)	64 (43-82)
Male sex, n (%)	89 (57)	70 (59)	31 (56)
ISS stage (I / II / III) at study entry, %[a]	40/31/25	34/30/30	36/25/33
High-risk cytogenetics, n (%)[b]	59 (38)	41 (34)	19 (35)
EMD at study entry, n (%)[c]	55 (35)	50 (42)	55 (100)
Median time since diagnosis (range), years	6.5 (0.7-24.6)	6.2 (0.7-24.6)	5.6 (0.7-14.4)
Median no. of prior lines of therapy (range)	5 (2-12)	5 (2-12)	5 (2-12)
Triple-class–refractory, n (%)[d]	119 (76)	119 (100)	50 (91)
Refractory to ≥1 anti-CD38 mAb	125 (80)	119 (100)	50 (91)
Refractory to prior alkylator therapy[e]	92 (59)	76 (64)	33 (60)

[a]At study entry, 4 patients had unknown ISS stage, and 2 patients had missing ISS stage in the ITT population.
[b]High-risk cytogenetics at study entry was based on fluorescence in situ hybridization defined as t(4;14), del(17/17p), and t(14;16) per Sonneveld P, et al[15]; 31 patients (20%) had unknown cytogenetics. Cytogenetic assessments were not centralized.
[c]EMD was defined as a multiple myeloma disease originating either in, but extending beyond, the cortical bone or as a separate soft tissue mass.
[d]Defined as refractory to or intolerant of ≥1 proteasome inhibitor, ≥1 immunomodulatory drug, and ≥1 anti-CD38 mAb.
[e]Including 21 patients (13%) refractory to prior melphalan in the ITT population.
EMD, extramedullary disease; ISS, International Staging System; ITT, intention-to-treat; mAb, monoclonal antibody.

- In the ITT population, median TTNT was 8.2 months (95% CI, 7.2-10.8), and median TTNT or death was 5.8 months (95% CI, 4.8-7.1) (**Figure 3**)
 - TTNTs of ≥3 months were achieved in 120 patients (76%), ≥6 months in 68 patients (43%), and ≥12 months in 12 patients (8%)
- Median TTNT and median TTNT or death in the triple-class–refractory population and in patients with EMD are shown in **Figure 4** and **Figure 5**, respectively

Figure 3. TTNT and PFS in the ITT Population (N=157)

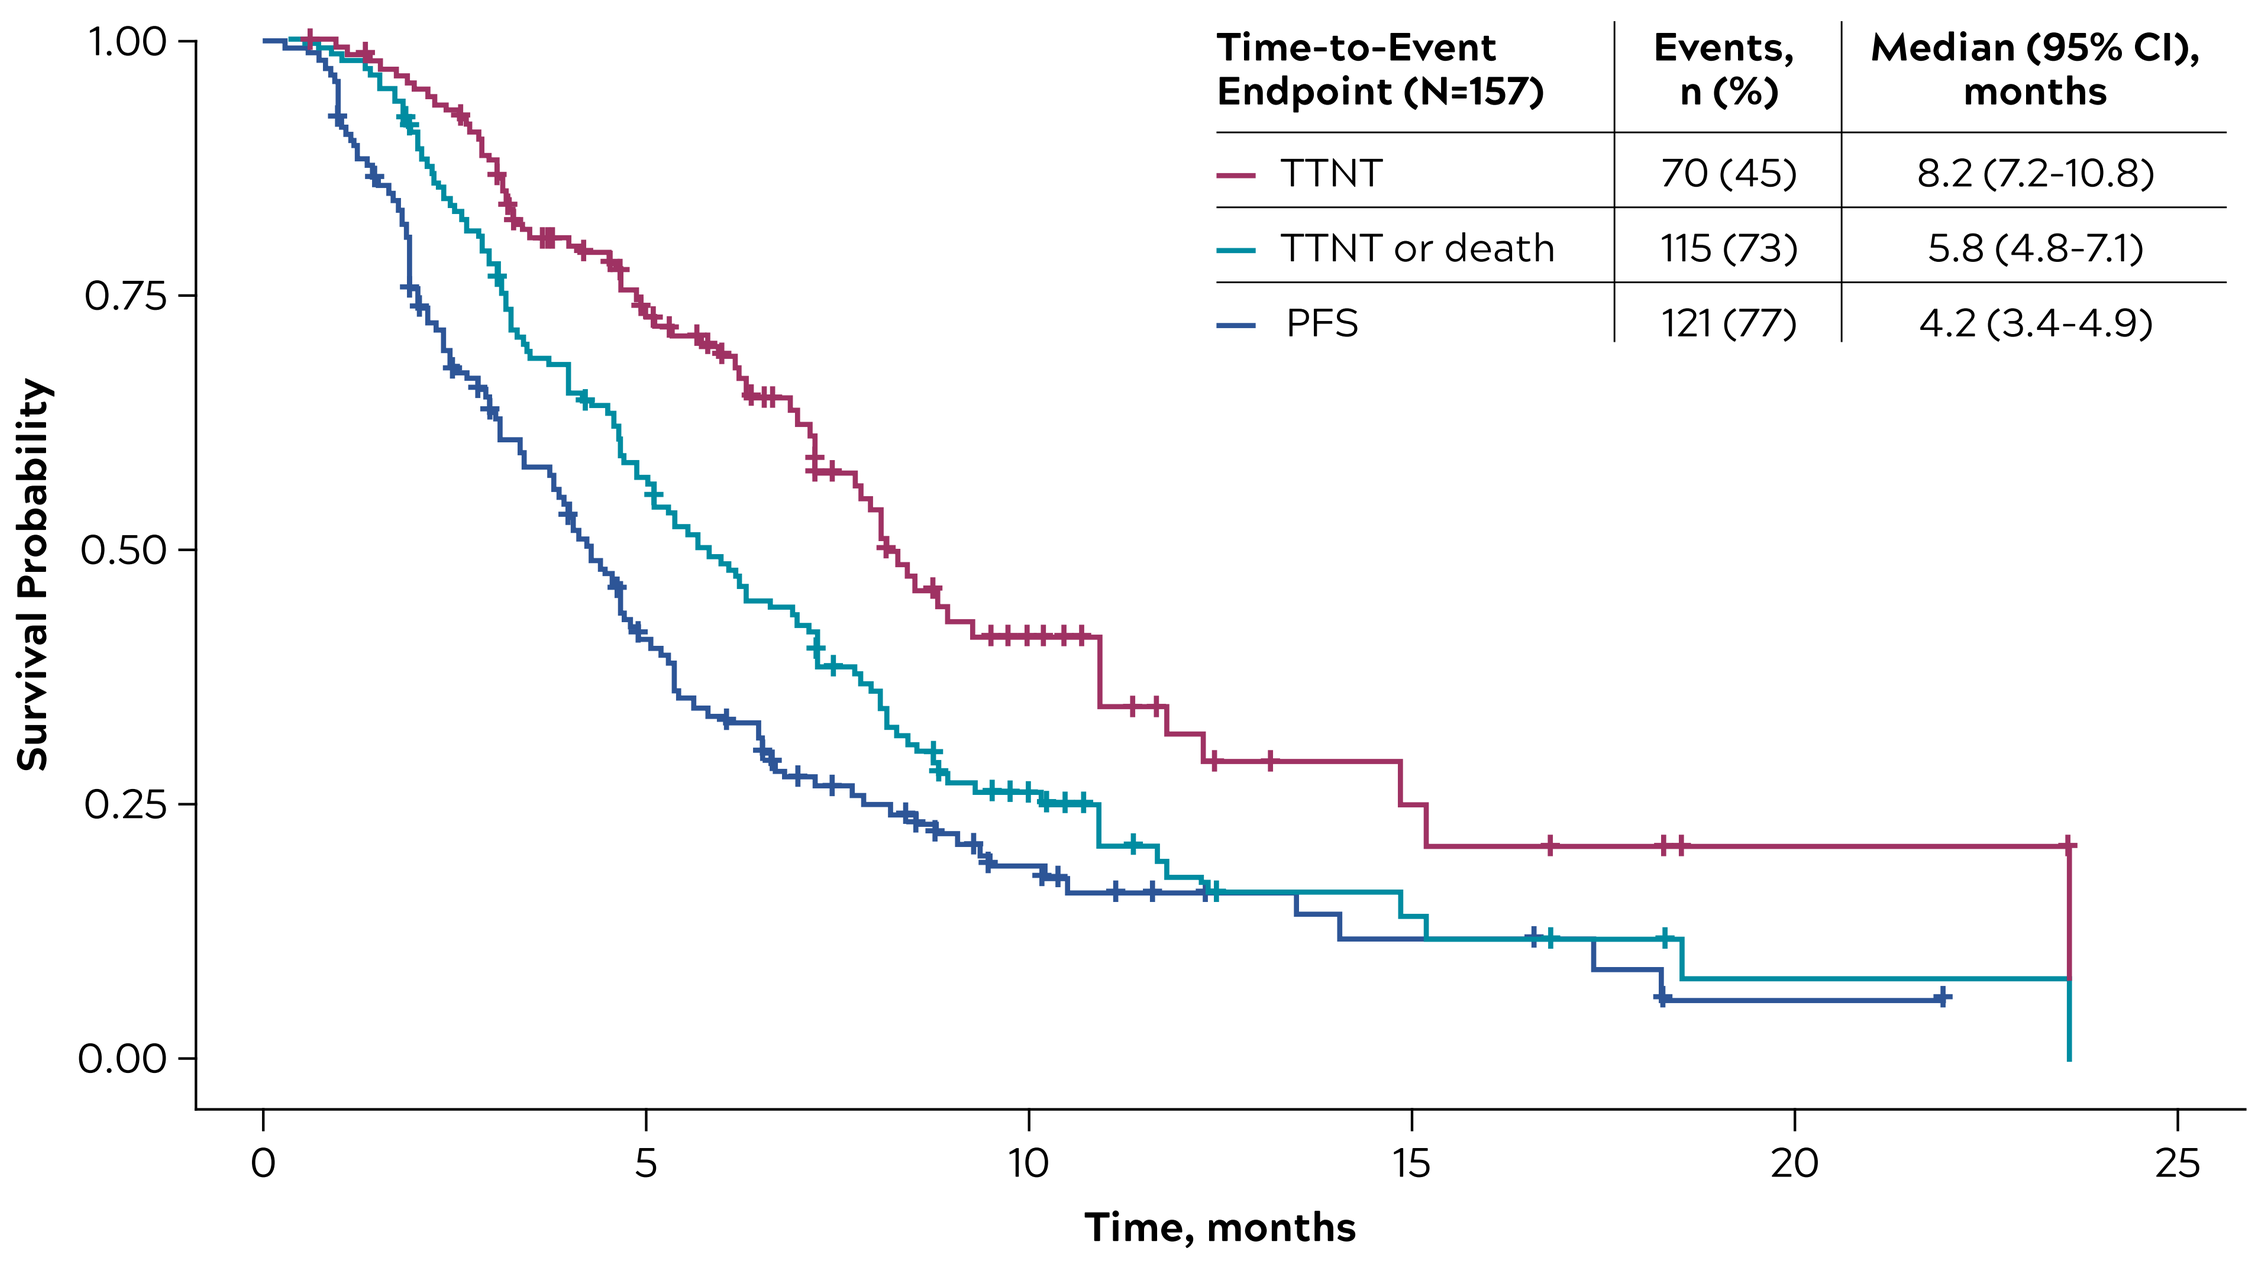
Survival Probability
1.00
0.75
0.50
0.25
0.00
0 5 10 15 20 25
Time, months
Time-to-Event Endpoint (N=157) | Events, n (%) | Median (95% CI), months
TTNT | 70 (45) | 8.2 (7.2-10.8)
TTNT or death | 115 (73) | 5.8 (4.8-7.1)
PFS | 121 (77) | 4.2 (3.4-4.9)

ITT, intention-to-treat; PFS, progression-free survival; TTNT, time to next treatment.

Figure 4. TTNT and PFS in the Triple-Class–Refractory Population (n=119)

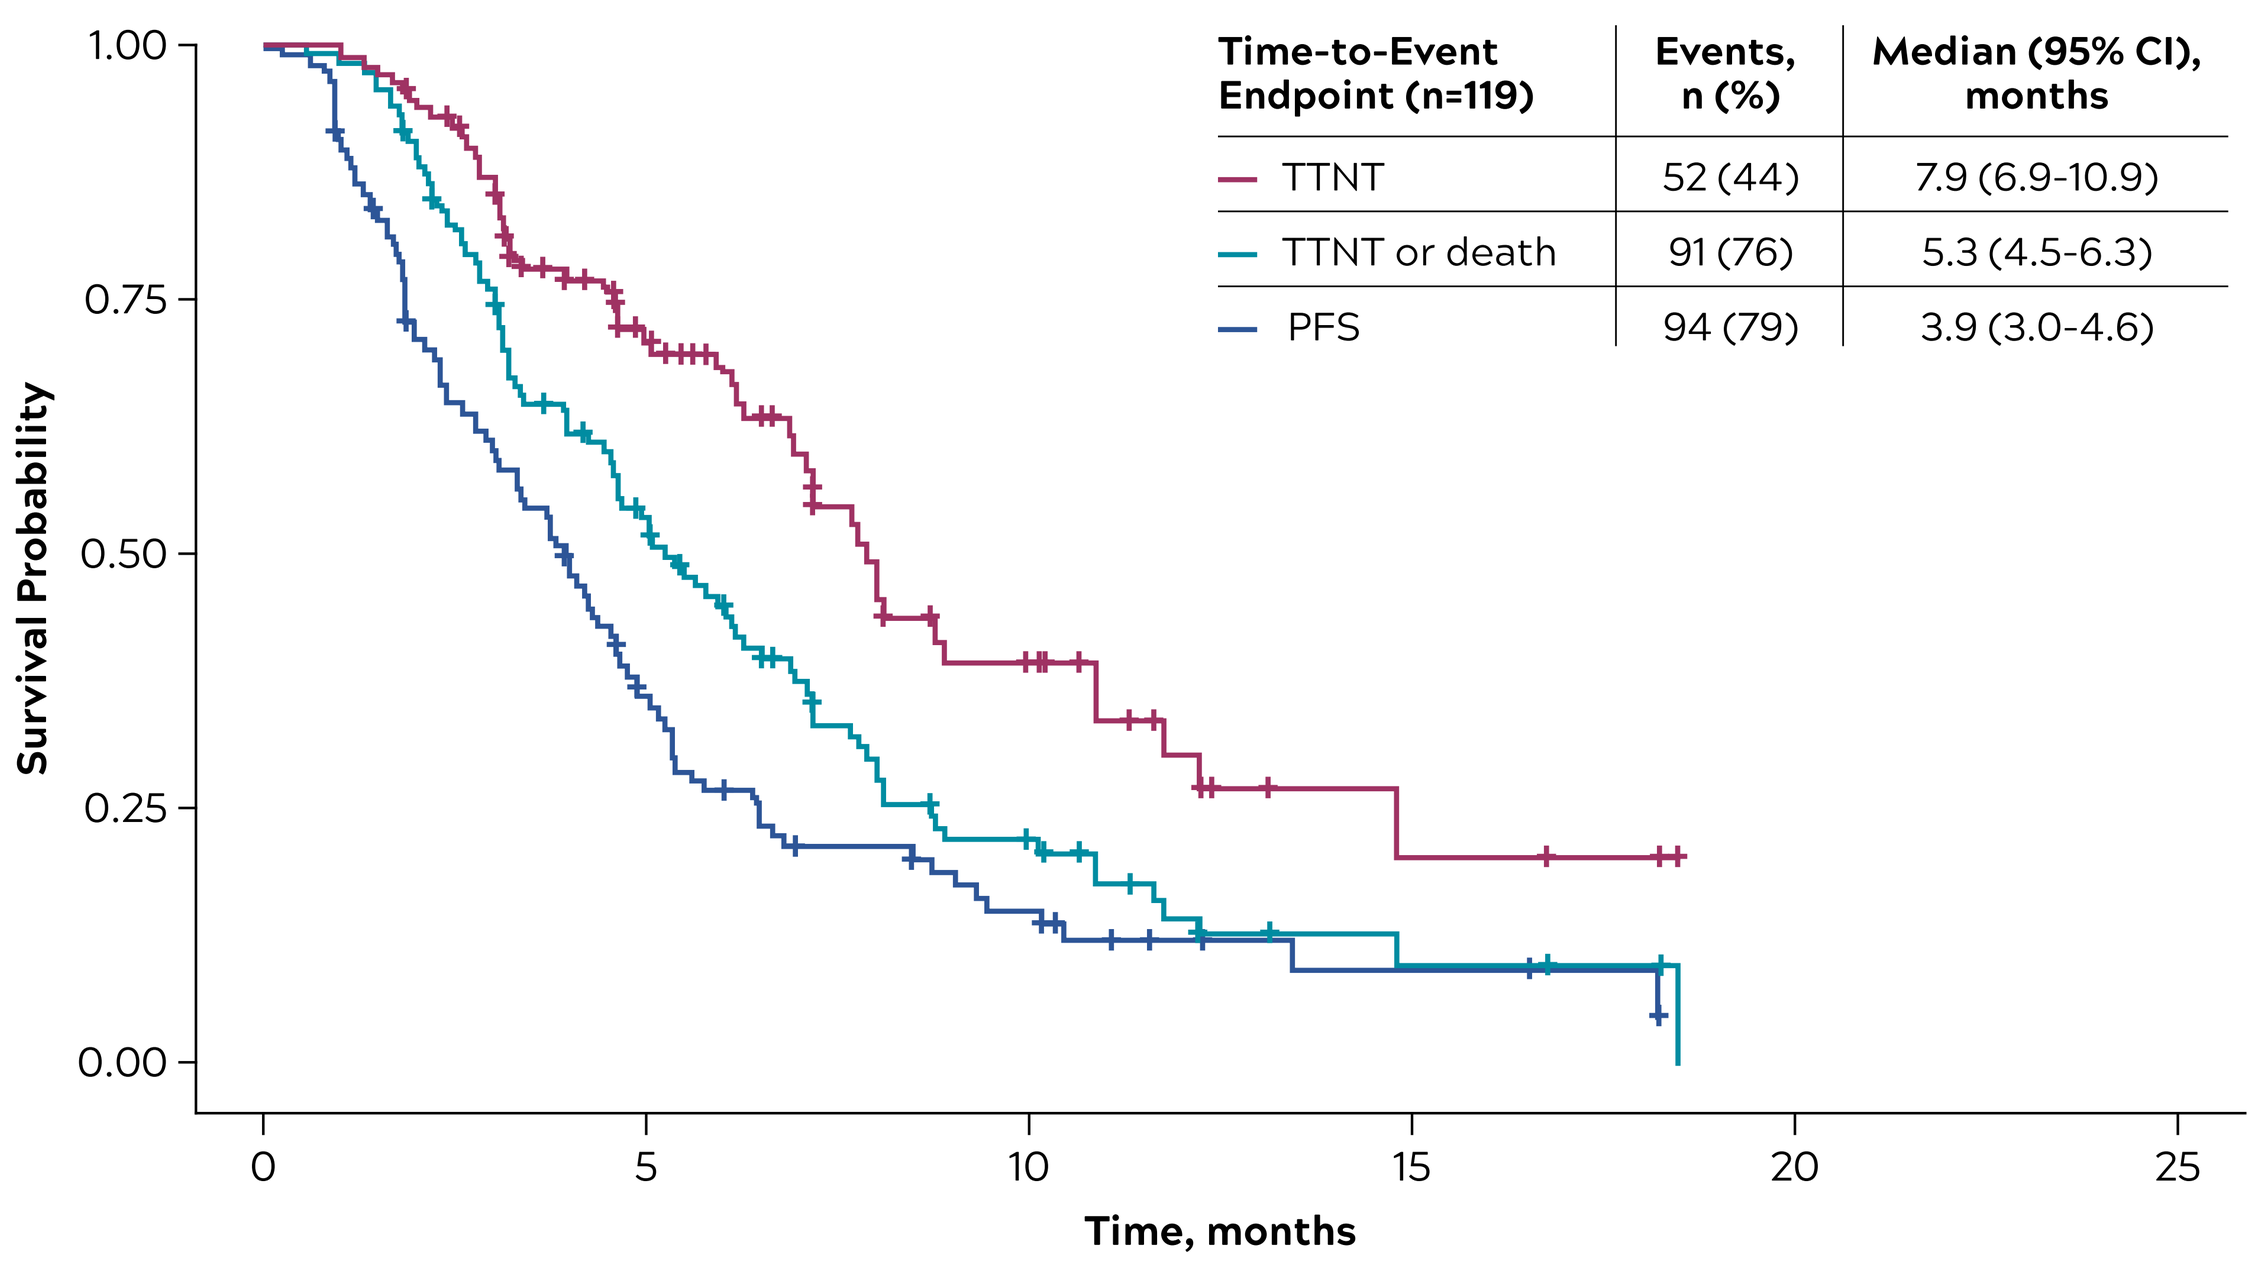
Survival Probability
1.00
0.75
0.50
0.25
0.00
0 5 10 15 20 25
Time, months
Time-to-Event Endpoint (n=119) | Events, n (%) | Median (95% CI), months
TTNT | 52 (44) | 7.9 (6.9-10.9)
TTNT or death | 91 (76) | 5.3 (4.5-6.3)
PFS | 94 (79) | 3.9 (3.0-4.6)

PFS, progression-free survival; TTNT, time to next treatment.

Figure 5. TTNT and PFS in Patients With EMD (n=55)

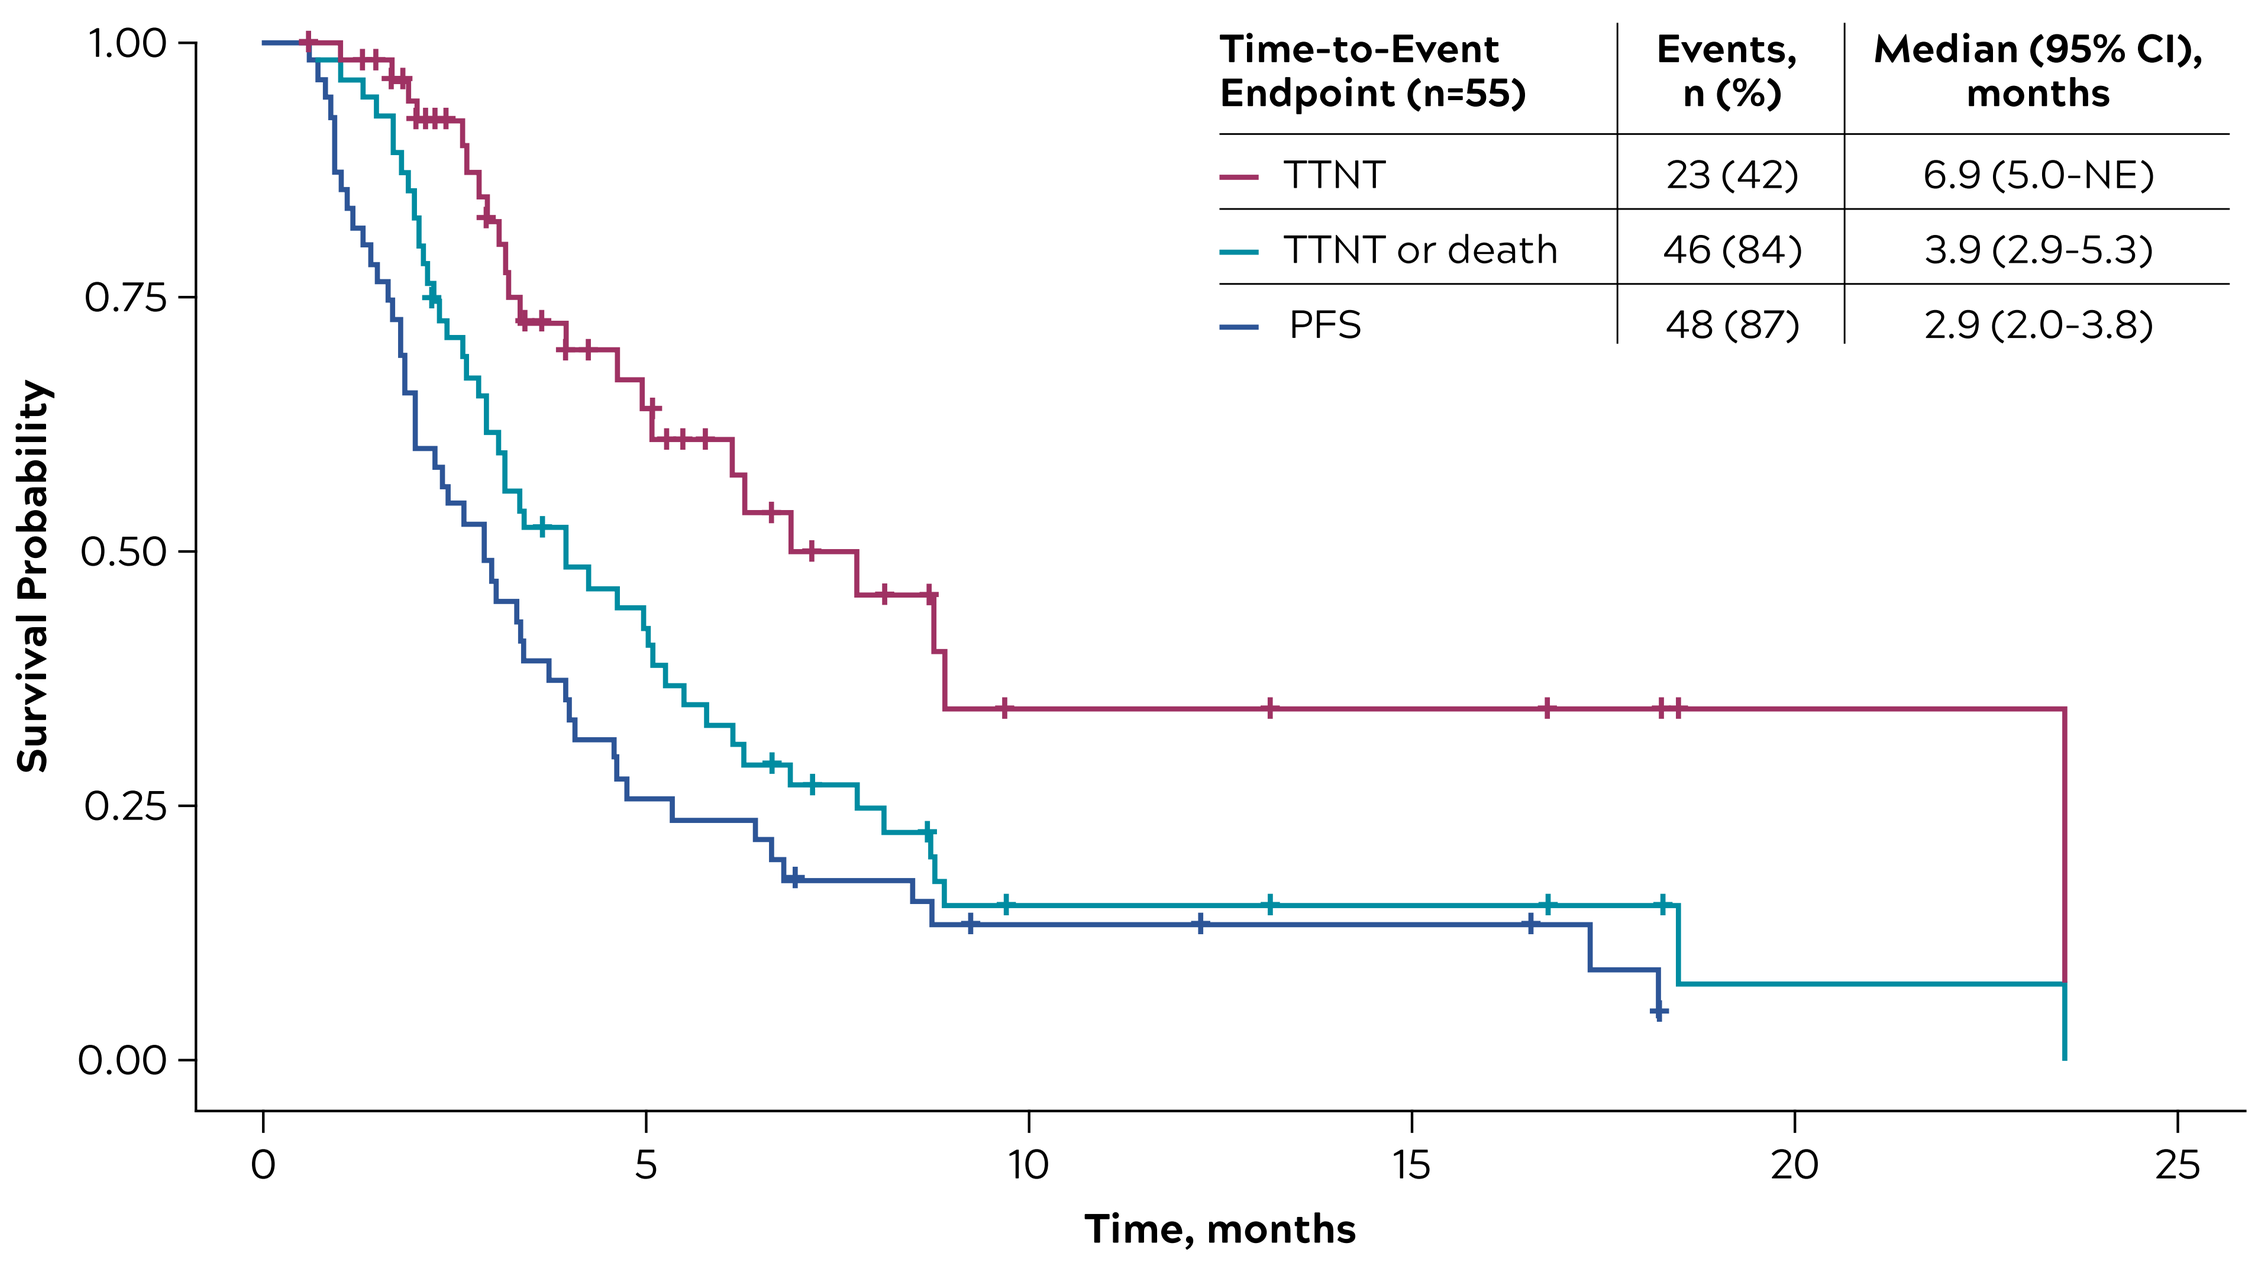
Survival Probability
1.00
0.75
0.50
0.25
0.00
0 5 10 15 20 25
Time, months
Time-to-Event Endpoint (n=55) | Events, n (%) | Median (95% CI), months
TTNT | 23 (42) | 6.9 (5.0-NE)
TTNT or death | 46 (84) | 3.9 (2.9-5.3)
PFS | 48 (87) | 2.9 (2.0-3.8)

EMD, extramedullary disease; NE, not estimable; PFS, progression-free survival; TTNT, time to next treatment.

- Of 131 patients who discontinued therapy, 70 patients (53%) initiated subsequent therapy at end of treatment
- Most patients received monotherapy as subsequent therapy, with antibodies and proteasome inhibitors being the most common types of therapies (**Table 3**)
- Patients had a high burden of disease; they had received multiple drug classes prior to the administration of melflufen and went on to receive a variety of treatments after melflufen (**Table 4**)
 - Results for patients with triple-class–refractory disease and EMD were consistent with those of the ITT population
 - Retreatment with the same drug class was generally uncommon in this heavily pretreated patient population, with retreatment with proteasome inhibitors being the most common (10 of 28 patients; 36%)

Table 3. Subsequent Treatment After Melflufen

Subsequent Therapy Received	ITT (n=70)	Triple-Class–Refractory (n=52)	EMD (n=23)
Therapy category, n (%)			
Monotherapy[a,b]	34 (49)	21 (40)	11 (48)
Doublet therapy[b]	27 (39)	24 (46)	11 (48)
≥3 Drug classes[b]	8 (11)	6 (12)	1 (4)
Therapy class, n (%)			
Antibody[c]	24 (34)	12 (23)	5 (22)
Anti-CD38-mAb	15 (21)	3 (6)	2 (9)
PI[d]	24 (34)	21 (40)	10 (44)
Alkylator[e]	18 (26)	15 (29)	7 (30)
IMiD[f]	16 (23)	15 (29)	6 (26)

[a]Excluding 1 patient who received dexamethasone monotherapy as subsequent therapy.
[b]Antimyeloma therapy with/without steroids.
[c]Included daratumumab, isatuximab, elotuzimab, nivolumab, and durvalumab.
[d]Included bortezomib, carfilzomib, and ixazomib.
[e]Included bendamustine, cyclophosphamide, and melphalan.
[f]Included lenalidomide, pomalidomide, and thalidomide.
EMD, extramedullary disease; ITT, intention-to-treat; mAb, monoclonal antibody; PI, proteasome inhibitor.

Table 4. Last Line of Therapy Prior to Melflufen and Subsequent Therapy After Treatment With Melflufen in the ITT Population

Last Regimen of Therapy Prior to HORIZON Study Start[a,b]	Subsequent Therapy After Melflufen, n (%)[a,b]		
	Monotherapy	Doublet Therapy	≥3 Drug Classes
Monotherapy (n=29)	17 (59)[c]	10 (34)	1 (3)
Doublet therapy (n=33)	14 (42)	13 (39)	6 (18)
≥3 Drug classes (n=8)	3 (38)	4 (50)	1 (13)

Last Line of Therapy Prior to HORIZON Study Start[a,b]	Subsequent Therapy After Melflufen, n (%)[a]				
	Antibody[d]	Anti-CD38 mAb	PI[e]	Alkylator[f]	IMiD[g]
Antibody (n=25)[d]	3 (12)	1 (4)	10 (40)	9 (36)	5 (20)
Anti-CD38 mAb (n=23)	2 (9)	0	9 (39)	9 (39)	5 (22)
PI (n=28)[e]	8 (29)	5 (18)	10 (36)	6 (21)	7 (25)
Alkylator (n=19)[f]	8 (42)	2 (11)	7 (37)	5 (26)	7 (37)
IMiD (n=32)[g]	15 (47)	12 (38)	15 (47)	5 (16)	6 (19)

[a]Among 70 patients who discontinued melflufen therapy in HORZON and went on to receive subsequent therapy.
[b]Antimyeloma therapy with/without steroids (eg, monotherapy: anti-CD38 mAb ± steroids; doublet therapy: anti-CD38 mAb + IMiD ± steroids).
[c]Excluding 1 patient who received dexamethasone monotherapy as subsequent therapy.
[d]Included daratumumab, isatuximab, elotuzimab, nivolumab, and durvalumab.
[e]Included bortezomib, carfilzomib, and ixazomib.
[f]Included bendamustine, cyclophosphamide, and melphalan.
[g]Included lenalidomide, pomalidomide, and thalidomide.
ITT, intention-to-treat; mAb, monoclonal antibody; PI, proteasome inhibitor.

- TTNT with melflufen plus dex is as good or better than that of other agents as presented in clinical trials and in real-world evidence studies in patients with RRMM (**Table 5**)

Table 5. TTNT With Melflufen and Other Agents in RRMM

Agent or Regimen	Study Type	Patients, n	Median Prior Lines of Therapy, n	Median TTNT, mo
Death censored				
Melflufen + dex (HORIZON)	Clinical trial	157	5	8.2
Melflufen + dex (O-12-M1)[6]	Clinical trial	45	4	10.6
Pom + dex[16]	Clinical trial	153	3	9.1
Multiple[4,a]	Retrospective	153/123	4/5	6/3
Death as an event				
Melflufen + dex (HORIZON)	Clinical trial	157	5	5.8
Melflufen + dex (O-12-M1)[6]	Clinical trial	45	4	7.9
Daratumumab[17]	Retrospective	126	4	5.7

[a]Retrospective study evaluating patients with RRMM treated from 2006-2016 at the Vejle Hospital. The 3 most common regimens patients received throughout the study, regardless of line of therapy, included lenalidomide + dex; bortezomib + dex; and bortezomib + lenalidomide + dex.[4]
Dex, dexamethasone; pom, pomalidomide; RRMM, relapsed/refractory multiple myeloma; TTNT, time to next treatment.

- The disease stabilization rate was 69% (95% CI, 61-76) in the ITT population, 65% (95% CI, 55-73) in the triple-class–refractory population, and 51% (95% CI, 37-65) in patients with EMD

CONCLUSIONS

- Melflufen plus dex treatment resulted in disease stabilization (≥SD) in 69% of patients, which translated to a median TTNT of 8.2 months (5.8 months when not censoring at time of death) in heavily pretreated patients with RRMM, including those with triple-class–refractory disease and EMD
- Results for TTNT from HORIZON (median, 5 prior lines) were consistent with previous reports of TTNT in patients with RRMM who received melflufen plus dex or other therapies (median, 3-4 prior lines). To our knowledge, this is the first report of TTNT from a trial population with such advanced RRMM
- In patients with RRMM, prolonged TTNT is associated with clinical benefit and health economic value for payors. Because TTNT results from clinical studies are not always applicable to the real-world setting, it is necessary that future clinical trials are more representative of the general RRMM population;[18-20] therefore, future studies of melflufen will gather real-world data
- Further analyses on TTNT and its relationship to health-related quality of life are being conducted and will be presented at a later date
- The high variability in subsequent therapies after melflufen plus dex is indicative of a lack of effective treatment options and a significant unmet medical need in patients with heavily pretreated RRMM
 - The proportion of patients who received subsequent therapies after melflufen plus dex is consistent with other reported data (53% vs 54%)[16]
- The efficacy and safety of melflufen is being further evaluated in OCEAN (OP-103), an ongoing, randomized, head-to-head, superiority, open-label, global, phase 3 study of melflufen plus dex vs pom plus dex in patients with RRMM refractory to lenalidomide (NCT03151811)

REFERENCES

1. Kumar SK, et al. *Leukemia*. 2017;31:2443-2448.
2. Gandhi UH, et al. *Leukemia*. 2019;33:2266-2275.
3. Yong K, et al. *Br J Haematol*. 2016;175:252-264.
4. Szabo AG, et al. *Clin Hematol Int*. 2019;1:220-228.
5. Chen CC, et al. *J Manag Care Spec Pharm*. 2017;23:236-246.
6. Bringhen S, et al. *J Clin Oncol*. 2019;37(suppl, abstr):8043.
7. Chauhan D, et al. *Clin Cancer Res*. 2013;19:3019-3031.
8. Wickström M, et al. *Invest New Drugs*. 2008;26:195-204.
9. Ray A, et al. *Br J Haematol*. 2016;174:397-409.
10. Strese S, et al. *Biochem Pharmacol*. 2013;86:888-895.
11. Wickström M, et al. *Oncotarget*. 2017;8:66641-66655.
12. Richardson PG, et al. Poster presented at the 25th EHA Annual Congress, June 11-21, 2020. Abstract EP945.
13. Richardson PG, et al. *Lancet Haematol*. 2020;7:e395-e407.
14. Rajkumar SV, et al. *Blood*. 2011;117:4691-4695.
15. Sonneveld P, et al. *Blood*. 2016;127:2955-2962.
16. Richardson PD, et al. Oral presentation at the 2019 ASCO Annual Meeting, May 31-June 4, 2019, Chicago, IL. Abstract 8004.
17. Lakshman A, et al. *Am J Hematol*. 2017;92:1146-1155.
18. Chari A, et al. *Blood*. 2017;130(suppl, abstr):1818.
19. Chari A, et al. *Expert Rev Hematol*. 2020;13:421-433.
20. Richardson PG, et al. *Blood Cancer J*. 2018;8:109.

ACKNOWLEDGMENTS

The authors would like to thank the patients who volunteered to participate in the study, the staff and the study sites who cared for them, and Precision for Medicine, the CRO involved in data gathering and analyses.
Medical writing support was provided by Katherine Mills-Lujan, PhD, CMPP, of Team 9 Science with funding from Oncopeptides.

DISCLOSURES

MVM received personal fees from AbbVie, Adaptive, Amgen, Celgene, GlaxoSmithKline, Janssen, Roche, Seattle Genetics, and Takeda. **AO** reports consultancy for Amgen, Celgene-Bristol-Myers Squibb, and Janssen. **AL** reports honoraria and personal fees from Bristol-Myers Squibb, Celgene, Janssen, and Takeda, and personal fees from Amgen and GlaxoSmithKline. **JB** reports honoraria from Amgen, Celgene, Janssen, Oncopeptides, and Takeda. **MC** received personal fees from AbbVie, Amgen, Bristol-Myers Squibb, Celgene, GlaxoSmithKline, Janssen, Sanofi, and Takeda. **PRO** reports honoraria from AbbVie, Amgen, Celgene-Bristol-Myers Squibb, GlaxoSmithKline, Janssen, Kite, Oncopeptides, and Sanofi. **HH** received grant support from Celgene and Janssen, and personal fees from Novartis. **CT** reports personal fees from Oncopeptides during the conduct of the study. **AA** received honoraria from Amgen, Bristol-Myers Squibb, Celgene, GlaxoSmithKline, Janssen, Oncopeptides, Sanofi, and Takeda. **AP** reports honoraria from Amgen and Celgene and consultancy/advisory fees from AbbVie, Cellectar, Janssen, Karyopharm, and Takeda. **CM** received honoraria from Amgen, Celgene, Incyte, Janssen, Karyopharm, Kite, Takeda, and Verastem, and consulting fees from Bayer. **XL, JWH, AM,** and **AR** have nothing to disclose. **JH** is a consultant of and owns stock and stock options in Oncopeptides. **SZ, ST,** and **OO** are employees of and own stock and stock options in Oncopeptides. **PGR** received grant support from Bristol-Myers Squibb, grant support and honoraria from Celgene, Karyopharm, Oncopeptides, and Takeda, and honoraria from Janssen, Sanofi, and SecuraBio.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Presented at the 6th World Congresses on Controversies in Multiple Myeloma (COMy), October 3-4, 2020

www. comy2020.cme-congresses.com